Reasons for Refusing Parenteral Therapy: A Qualitative Study of Patients with Pulmonary Arterial Hypertension

Morland K[1]; Nails A[1]; Classi P[1]; Morreall J[2]; Etschmaier M[2]

[1]United Therapeutics Corporation, Durham, NC; [2]Xcenda LLC, Tampa, FL

BACKGROUND

- The prostacyclin pathway is an important target in the treatment of pulmonary arterial hypertension (PAH).
- Parenteral prostacyclin therapy is the treatment of choice for PAH patients with advanced disease; it can also be used as part of combination therapy in patients with less severe disease who do not respond to monotherapy.[1,2]
- Despite a clear clinical benefit and guidelines supporting its use, many patients never receive parenteral prostacyclin therapy prior to death.[3-5]
- In the REVEAL registry, only 43% of patients who died during the study period were receiving IV prostacyclin as monotherapy or combination therapy prior to death due to any cause.[3]
- A chart review of PAH patients being treated at a large pulmonary hypertension center found most patients who died without parenteral prostacyclin therapy (N=40) were not considered candidates (60%), refused parenteral prostacyclin therapy (15%), or had no documented evaluation for parenteral prostacyclin therapy (25%).[4]
- This study sought to understand, directly from the PAH patient, why they refused parenteral prostacyclin therapy and, in some cases, later reversed that decision.

OBJECTIVES

- To gain an in depth understanding of the reasons why patients decline parenteral prostacyclin therapy and what consequences may arise from that decision.
- To understand why some patients who initially refused parenteral prostacyclin therapy later reversed that decision and their subsequent experience.
- To gain insight into new pump/device features that may impact a patient's decision to accept treatment with a parenteral prostacyclin therapy.

METHODS

- Double-blinded, qualitative, 45-minute telephone interviews were conducted with PAH patients who refused parenteral prostacyclin therapy.
- The study received approval from Advarra Institutional Review Board prior to initiating recruitment.
- Rare Patient Voice®, a rare and orphan disease survey and interview company, used their internal database of patients who have opted in to participate in qualitative research studies to recruit PAH patients via email invitation to complete a screening questionnaire. Eligible patients were ≥18 years of age, diagnosed with PAH, and were recommended parenteral prostacyclin therapy in the previous 2 years.
- Patients answered a series of questions designed to assess their decision-making process when asked to consider parenteral prostacyclin therapy, including treatment perceptions and consequences that occurred as a result of refusing therapy. In those who subsequently initiated parenteral prostacyclin therapy, patients were asked to compare their perceptions to their actual experience. Finally, patient perceptions of potential new pump/device features were discussed and rated.
- Respondents were evaluated overall and divided into two groups for analysis: those who refused parenteral prostacyclin therapy and never initiated it (Group A) and those who initially refused parenteral prostacyclin therapy but subsequently initiated it (Group B). Both quantitative and qualitative analyses were performed on the data.

RESULTS

- Of the 754 patients who were contacted, 92 passed screening criteria and 25 completed the interview.
- 36% (n=9) and 64% (n=16) of patients interviewed were in Groups A and B, respectively. Patients in the overall sample (N=25) had a mean [SD] age of 47.2 [9.2], were primarily female (88%), and had been diagnosed with PAH for 9.9 [8.7] years.

Table 1. Patient Demographics

	All Patients (N=25)	Group A* (N=9)	Group B* (N=16)
Age in years, mean (SD)	47.2 (9.2)	52.0 (5.7)	44.4 (9.8)
Female, n (%)	22 (88)	8 (89)	14 (88)
Years since PAH diagnosis, mean (SD)	9.9 (8.7)	4.4 (2.8)	13.0 (9.4)
Education Level, n (%)			
High school graduate	2 (8)	1 (11)	1 (6)
Trade or vocational school graduate	1 (4)	1 (11)	0 (0)
Some college	6 (24)	1 (11)	5 (31)
Associate's degree	3 (12)	1 (11)	2 (13)
Bachelor's degree	9 (36)	4 (44)	5 (31)
Graduate degree	4 (16)	1 (11)	3 (19)
Ethnicity, n (%)			
White	19 (76)	8 (89)	11 (69)
Black/African American	2 (8)	0 (0)	2 (13)
Not specified/Other	3 (12)	1 (11)	2 (13)
Hispanic	1 (4)	0 (0)	1 (6)
Relationship status, n (%)			
Married/significant other	12 (48)	6 (67)	6 (37)
Single/divorced/widowed	13 (52)	3 (33)	10 (63)
Income, n (%)			
<$10,000	2 (8)	1 (11)	1 (6)
$10,000-$25,000	4 (16)	0 (0)	4 (25)
$26,000-$50,000	8 (32)	2 (22)	6 (37)
$51,000-$100,000	9 (36)	5 (56)	4 (25)
$101,000-$200,000	1 (4)	1 (11)	0 (0)
>$200,000	1 (4)	0 (0)	1 (6)

*Group A included those patients who refused parenteral prostacyclin therapy and never initiated it. Group B included those patients who initially refused parenteral prostacyclin therapy but subsequently initiated it.

Decision-making Process Upon Initial Consideration of Parenteral Prostacyclin Therapy

- Most patients reported being assessed as having severe disease by their healthcare provider at initial diagnosis (78% in Group A and 69% in Group B, respectively).
- At the time their healthcare providers suggested parenteral prostacyclin therapy, patients most commonly reported being told it was necessary due to their pressures/echo findings, the severity of their disease, and that it's the most direct/consistent therapy given the continuous IV infusion. Providers shared information about the positive and negative effects of therapy, informing patients of side effects and potential risks of having a central line while characterizing the potential benefits as improving symptoms, functionality, and disease progression.
- When describing their initial reaction when asked to consider parenteral prostacyclin therapy, patients frequently used terms like *"fear," "permanent," "interfere,"* and *"being attached."*
- Patients consulted family and friends, their medical providers, and support groups (eg, Pulmonary Hypertension Association) when considering pump therapy. Approximately half of patients stated they reached out to other users of pump therapy online and through support groups to understand their experience.
- Specific topics patients researched included medication side effects, therapy and pump options, financial resources to pay for therapies, general disease information and infection risk/central line information.

Patient Perceptions of Parenteral Prostacyclin Therapies

- In both Groups, patients expressed an expectation that parenteral prostacyclin therapy would improve their symptoms and positively impact their disease progression; however, they frequently expressed fear and concern about the impact of parenteral prostacyclin therapy on their daily life as it relates to wearing an external pump for an interminable amount of time, an inability to swim/challenges showering, administering the medication, the potential for line infections for intravenous use and site pain for subcutaneous use, and their ability to work or care for children.

Comparison of Perception to Experience

- Among patients who eventually initiated parenteral prostacyclin therapy (Group B), the impact of treatment on symptoms, disease progression, and survival was aligned with their perception prior to initiation. Patients anticipated the occurrence of common side effects associated with parenteral prostacyclin therapy, including subcutaneous site-pain and line-related issues, and stated their experience was as expected. Among those patients who perceived managing/physically wearing the pump to negatively impact their quality of life, they characterize their experience as *"less of a big deal," "a new routine,"* and you *"learn to live with it."*

Primary Reason for Treatment Decision

- Patients who ultimately initiated parenteral prostacyclin therapy (Group B) most frequently stated they did so due to a worsening of symptoms, disease progression, or a desire to live.
 - *"I [had] spoken to my parents and close relatives or friends. And everyone, of course, wanted me to go on it because that would keep me alive. But it was my decision ultimately, of course, and I've had enough."*
- Patients who never initiated parenteral prostacyclin therapy (Group A) characterized their values as a balance between quality of life and prolonging life. Most would reconsider PAR if their disease started to progress rapidly or if they felt there were no other options.
 - *"I told my doctor, I said if it progresses the opposite direction and we're not moving forward, then I will definitely consider doing that because I do want to live as long as I can but I want the best quality of that time."*

Consequences of Declining Parenteral Prostacyclin Therapy

- When asked whether they experienced unplanned hospitalizations or emergency room (ER) visits since declining pump therapy, patients had a mixed experience. Some experienced no unplanned visits, while others did, both related and unrelated to their PAH.

Perceptions of New Pump/Device Features

- The new pump/device features patients reported to be the most impactful to increasing their acceptance of parenteral prostacyclin therapy included water resistance, a pre-filled pump, implantability, and smaller size.
- The ability to have a pre-filled pump was rated the most impactful by Group A while water resistance was rated the most impactful by Group B.
- When asked which pump feature(s) would change your mind about declining a parenteral prostacyclin therapy, the most frequently mentioned features were implantability (60%) followed by water resistance (36%).

Figure 1. Impact of New Pump/Device Features on Patients' Interest in Using a Parenteral Prostacyclin Therapy

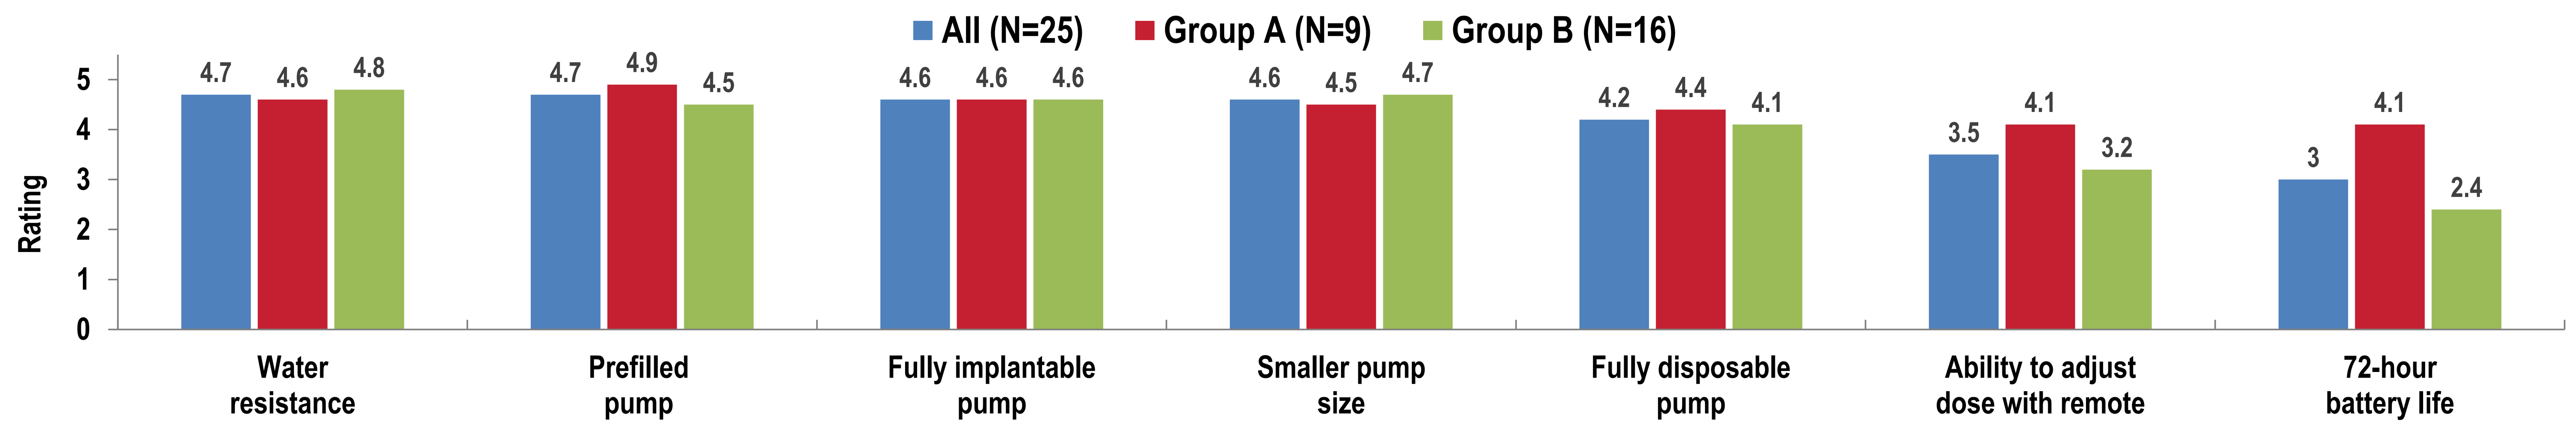
All (N=25)
Group A (N=9)
Group B (N=16)
Rating
0
1
2
3
4
5
4.7 4.6 4.8
4.7 4.9 4.5
4.6 4.6 4.6
4.6 4.5 4.7
4.2 4.4 4.1
3.5 4.1 3.2
3 4.1 2.4
Water resistance
Prefilled pump
Fully implantable pump
Smaller pump size
Fully disposable pump
Ability to adjust dose with remote
72-hour battery life

LIMITATIONS

- As with any patient-reported research, this study is potentially subject to selection bias, as patients volunteered to participate in the interview if they met the screening criteria.
- Due to the small sample size, the patients in this study may not be fully representative of the broader PAH population considered candidates for parenteral prostacyclin therapy.
- The small sample size may limit our ability to identify differences (if any) between those patients who refused parenteral prostacyclin therapy and never initiated it vs those who initially refused parenteral prostacyclin therapy but subsequently initiated it.

CONCLUSIONS

- This is the first study to our knowledge to explore, directly from the patient, why they decline parenteral prostacyclin therapy.
- Qualitative analysis shows that while patients understand the potential benefit of parenteral prostacyclin therapy on improving their disease, they fear the impact of using and managing a pump modality on their quality of life, and therefore decline treatment.
- Additional considerations should be given to overcome patients' expressed concerns, so they do not delay treatment with parenteral prostacyclin therapy when deemed appropriate.
- Enhancements in pump technology would likely increase acceptance and reduce the delay to parenteral prostacyclin therapy for appropriate patients.

REFERENCES

1. Galie N, Humbert M, Vachiery JL, et al. 2015 ESC/ERS Guidelines for the diagnosis and treatment of pulmonary hypertension: The Joint Task Force for the Diagnosis and Treatment of Pulmonary Hypertension of the European Society of Cardiology (ESC) and the European Respiratory Society (ERS): Endorsed by: Association for European Paediatric and Congenital Cardiology (AEPC), International Society for Heart and Lung Transplantation (ISHLT). *Eur Respir J* 2015; 46: 903-975.
2. McLaughlin VV, Archer SL, Badesch DB, et al. ACCF/AHA 2009 expert consensus document on pulmonary hypertension a report of the American College of Cardiology Foundation Task Force on Expert Consensus Documents and the American Heart Association developed in collaboration with the American College of Chest Physicians' American Thoracic Society, Inc.; and the Pulmonary Hypertension Association. *J Am Coll Cardiol*. 2009;53:1573-619.
3. Farber HW, Miller DP, Meltzer LA, McGoon MD. Treatment of patients with pulmonary arterial hypertension at the time of death or deterioration to functional class IV: Insights from the REVEAL Registry. *J Heart Lung Transplant*. 2013;32:1114-1122.
4. Hay BR, Pugh ME, Robbins IM, Hemnes AR. Parenteral prostanoid use at a tertiary referral center: A retrospective cohort study. *Chest* 2016;149(3):660-666.
5. Tonelli AR, Arelli V, Minai OA, et al. Causes and circumstances of death in pulmonary arterial hypertension. *Am J Resp Crit Care Med*. 2013;188(3):365-369.

Presented at CHEST 2020 Virtual | P1879

Disclosures: This study was sponsored by United Therapeutics Corporation.

United Therapeutics CORPORATION